A MULTIPLE-DOSE STUDY OF BLOCKADE OF OPIOID SUBJECTIVE EFFECTS BY SUBCUTANEOUS INJECTIONS OF DEPOT BUPRENORPHINE IN SUBJECTS WITH OPIOID USE DISORDER

H. Jia[1], M.K. Greenwald[2], B. D. Vince[3], P. J. Fudala[1], C. Heidbreder[1], A. F. Nasser[1];

[1]Indivior PLC, Richmond, VA, [2]Wayne State University, Detroit, MI, [3]Vince & Associates Clinical Research, Inc., Overland Park, KS

BACKGROUND

RBP-6000 is a 28-day SC injection of depot buprenorphine (Bup). We hypothesized (Nasser et al., 2014, *Clin Pharmacokinet*) that brain µ-opioid receptor occupancy (µORO) ≥ 70% and Bup plasma levels ≥ 2 ng/mL are needed to provide full blockade of opioid agonist effects. This Phase 2 study was designed to assess hydromorphone (HM) subjective and reinforcing effects in non treatment-seeking individuals presenting with moderate or severe opioid use disorder according to DSM-V.

METHODS

Thirty-nine subjects with opioid use disorder first completed 3 HM challenges [0, 6, 18 mg intramuscular (IM)] on 3 consecutive days in randomized order), then 3 HM challenges at the end of 14-day SUBOXONE film stabilization. This was followed by two RBP-6000 300-mg SC doses separated by 28 days. For 12 weeks after the first RBP-6000 dose, on days 5-7 of each week, subjects received 3 HM challenges in randomized order (6 sequences).

A Drug Liking visual analog scale (VAS) score was the primary endpoint, and HM reinforcing efficacy ($\log_{10}$ breakpoint values), and Any Effects, Bad Effects, High, Good Effects, and Sedation VAS scores were secondary endpoints. Statistical comparison using mixed effects model was used for each week. Change from HM 0 mg with 95% CI was reported, with a VAS difference cut-off of ≤11 required to declare full blockade. A blood sample for PK analysis was collected the morning of each HM administration day. Our published E_{max} model was used to calculate µORO.

VAS scores were collected at 30 min before and 15, 30, 45, 60, 75, 90, 120, 150, 180, 210, 240, 270, and 300 min after receiving the daily HM challenge. HM reinforcing efficacy was measured at least 5 hours after the daily HM challenge. Subjects completed a 12-trial choice session during which they could work (on a progressive ratio schedule) for units of the total amount of HM dosed earlier that day or for $2 units of money. The purpose of the choice task was to evaluate the reinforcing efficacy of the daily randomized HM challenge dose relative to money.

STUDY DESIGN

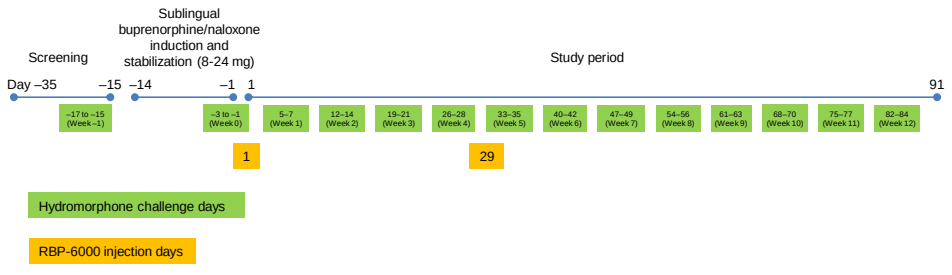
Screening
Sublingual buprenorphine/naloxone induction and stabilization (8-24 mg)
Study period
Day −35
−15
−14
−1
1
91
−17 to −15 (Week −1)
−3 to −1 (Week 0)
5–7 (Week 1)
12–14 (Week 2)
19–21 (Week 3)
26–28 (Week 4)
33–35 (Week 5)
40–42 (Week 6)
47–49 (Week 7)
54–56 (Week 8)
61–63 (Week 9)
68–70 (Week 10)
75–77 (Week 11)
82–84 (Week 12)
1
29
Hydromorphone challenge days
RBP-6000 injection days

BLOCKADE OF HM 6 MG CHALLENGE DOSE

Phase	Week	Bup plasma concen-tration (ng/mL)	Estimated brain µORO (%)	Drug Liking effect (VAS scores): change from placebo, LS mean (95% CI)	Reinforcing effect (breakpoint): change from placebo, LS mean of log transformation (95% CI)
		6 mg	6 mg	6 mg vs. placebo	6 mg vs. placebo
Screening	−1	0.00	0.00	45.36 (37.16, 53.56)	2.14 (1.55, 2.72)
Sublingual	0	1.32	54.64	8.20 (1.47, 14.94)	1.33 (0.45, 2.22)
RBP-6000	1	2.16	67.88	3.66 (−0.03, 7.34)	0.61 (−0.003, 1.23)
	2	1.87	66.06	0.59 (−1.98, 3.15)	0.74 (−0.12, 1.61)
	3	1.88	66.00	0.86 (−3.05, 4.78)	0.95 (0.31, 1.58)
	4	1.78	65.11	3.32 (−1.43, 8.06)	1.39 (0.71, 2.07)
	5	3.65	76.29	0.74 (−0.94, 2.42)	−0.09 (−1.09, 0.90)
	6	3.53	75.70	0.35 (−3.62, 4.32)	0.12 (−0.94, 1.17)
	7	3.50	75.64	−0.15 (−2.16, 1.86)	0.72 (−0.01, 1.44)
	8	3.37	74.79	−1.05 (−4.77, 2.68)	0.52 (−0.38, 1.41)
	9	3.12	73.97	−0.12 (−6.20, 5.96)	0.09 (−1.41, 1.59)
	10	3.04	73.08	−0.09 (−0.69, 0.51)	0.03 (−0.94, 1.01)
	11	2.99	73.05	−0.32 (−3.97, 3.34)	−0.22 (−1.69, 1.25)
	12	2.65	71.31	−0.03 (−2.19, 2.12)	0.59 (−0.57, 1.76)

BLOCKADE OF HM 18 MG CHALLENGE DOSE

Phase	Week	Bup plasma concen-tration (ng/mL)	Estimated brain µORO (%)	Drug Liking effect (VAS scores): change from placebo, LS mean (95% CI)	Reinforcing effect (breakpoint): change from placebo, LS mean of log transformation (95% CI)
		18 mg	18 mg	18 mg vs. placebo	18 mg vs. placebo
Screening	−1	0.00	0.00	60.61 (52.32, 68.90)	2.06 (1.48, 2.64)
Sublingual	0	1.24	53.63	17.17 (10.43, 23.90)	1.93 (1.10, 2.76)
RBP-6000	1	2.04	67.09	6.93 (3.24, 10.61)	0.94 (0.35, 1.53)
	2	1.97	66.49	2.90 (0.33, 5.47)	1.46 (0.65, 2.27)
	3	1.92	66.44	4.93 (1.02, 8.84)	0.91 (0.29, 1.52)
	4	1.81	65.35	6.68 (1.94, 11.42)	1.89 (1.23, 2.55)
	5	3.67	76.42	1.21 (−0.43, 2.85)	0.37 (−0.55, 1.30)
	6	3.52	75.69	3.16 (−0.83, 7.15)	0.98 (−0.07, 2.02)
	7	3.47	75.43	1.88 (−0.11, 3.87)	0.87 (0.10, 1.65)
	8	3.50	75.18	1.93 (−1.79, 5.66)	0.85 (−0.11, 1.81)
	9	3.21	74.04	4.17 (−1.84, 10.17)	1.10 (−0.31, 2.50)
	10	3.19	74.18	0.13 (−0.50, 0.76)	0.45 (−0.50, 1.41)
	11	3.08	73.51	3.24 (−0.35, 6.83)	0.75 (−0.66, 2.15)
	12	2.62	71.30	2.78 (0.61, 4.96)	1.59 (0.50, 2.67)

MEAN VAS SCORES FOLLOWING 6 & 18 MG HM CHALLENGE DOSES

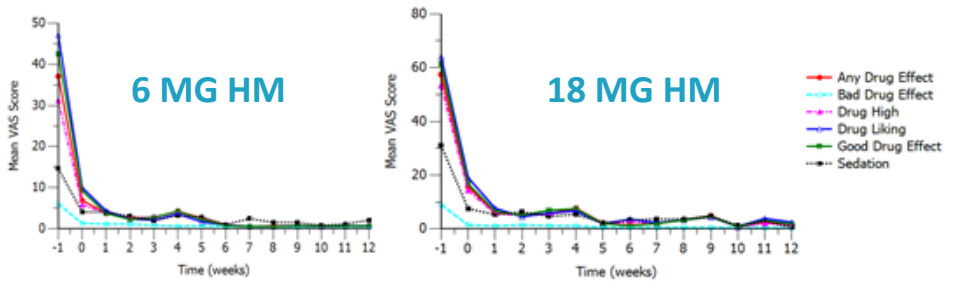
6 MG HM
18 MG HM
Mean VAS Score
Time (weeks)
Any Drug Effect
Bad Drug Effect
Drug High
Drug Liking
Good Drug Effect
Sedation

RBP-6000 blocked the subjective drug effects of HM.

"DRUG LIKING" & BREAKPOINT BY HM CHALLENGE DOSE

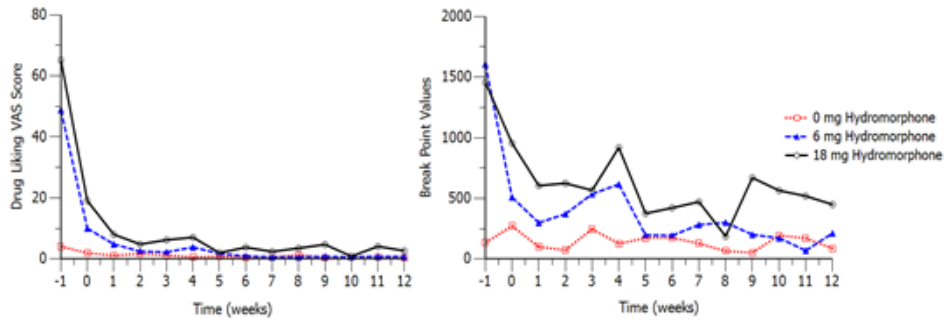
Drug Liking VAS Score
Break Point Values
Time (weeks)
0 mg Hydromorphone
6 mg Hydromorphone
18 mg Hydromorphone

RBP-6000 reduced HM Drug Liking and HM breakpoint.

CORRELATION BETWEEN BUP CONC & CLINICAL EFFECT

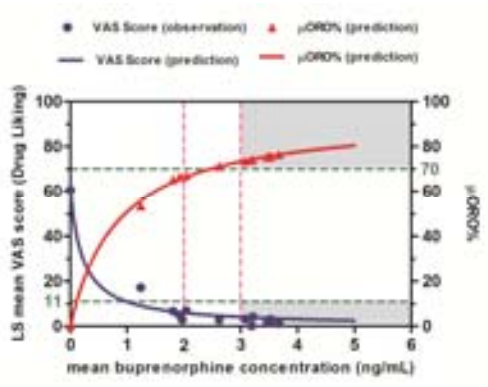
VAS Score (observation)
µORO% (prediction)
VAS Score (prediction)
µORO% (prediction)
LS mean VAS score (Drug Liking)
µORO%
mean buprenorphine concentration (ng/mL)

Red curve: Relationship between predicted µORO% and Bup plasma concentration (Nasser et al., 2014, *Clin Pharmacokinet*).

Blue curve: E_{max} inhibitory model predicts the relationship between Drug Liking VAS scores and Bup plasma concentration after 18 mg HM challenge.

Parameter	Units	Estimate	Std Error	CV%
E0	-	60.71	2.99	4.92
IC50	ng/mL	0.22	0.04	18.45

$$E = E0 * (1 - \frac{C}{C + IC50})$$

IC_{50} is the Bup plasma concentration that results in half of the E_0 effect.

•At baseline (week -1), mean Bup plasma concentrations were 0 ng/mL and the mean Drug Liking, Any Effects, Bad Effects, High, Good Effects and Sedation VAS scores were between 30 to 65 and the mean raw breakpoint values were approximately 1500 after 18 mg and 6 mg HM challenges.

•During the sublingual stabilization phase (median SL dose: 16 mg, range: 8 – 24 mg), mean Bup plasma concentrations at steady state were on average of 1.2 ng/mL, estimated µOROs were approximately 54% with associated high VAS scores (95% CI: 10.4-23.9) which did not demonstrate full opioid blockade (upper limit of CI exceeded 11 units).

MEAN AND 95%CI "DRUG LIKING" VAS SCORES BY WEEK

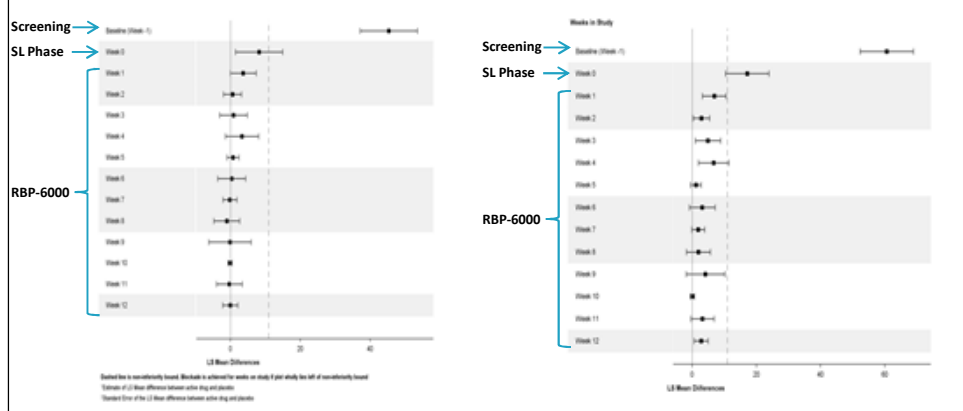
6 MG HM VS PLACEBO
18 MG HM VS PLACEBO
Screening
SL Phase
RBP-6000
Screening
SL Phase
RBP-6000

•After the 1st SC injection, mean Bup plasma concentrations were 1.8-2 ng/mL and µOROs were 65-68%. The mean Drug Liking VAS scores were 2.9-7 (<11, non-inferiority bound) and the mean breakpoint values were 0.9-1.9 following the 18 mg HM challenge.

•After the 2nd SC injection, Bup plasma concentrations were 2.6-3.7 ng/mL and µOROs were all >70%. The mean Drug Liking VAS scores were 0.1-4.2 and the mean breakpoint values were 0.3-1.6 with both 95% CIs including 0 after 18 mg HM challenge, confirming that our hypothesis (Nasser et al., 2014, *Clin Pharmacokinet*) was correct.

•The mean VAS scores and breakpoints increased at the end of the 1st SC injection (weeks 4) and at the end of the 2nd SC injection (week 9), which were related to the slight decrease in Bup concentrations, confirming that our 28-day dosing interval is appropriate.

•E_{max} inhibitory model with IC_{50} = 0.22 ng/mL described the relationship between Drug Liking VAS scores and Bup plasma concentration.

•Linear model with y = -0.4079x + 2.179 (R^2=0.6459) described the relationship between reinforcement efficacy breakpoint values and Bup plasma concentration.

CONCLUSIONS

- Findings from this study confirm the hypothesis that µORO ≥ 70% and Bup plasma levels ≥ 2 ng/mL are needed to provide full blockade of opioid agonist effects.
- The first dose of RBP-6000 (300 mg) (within the first week) was non-inferior to sublingual buprenorphine/naloxone in blocking the subjective and reinforcing effects of HM in subjects with moderate or severe opioid use disorder.
- RBP-6000 (300 mg) effectively blocked HM's subjective effects and reinforcing efficacy suggesting that treatment with RBP-6000 may significantly reduce illicit opioid use. It is hoped that RBP-6000 will be useful in patients with moderate or severe opioid use disorder, and may improve patient adherence (monthly dosing) and reduce diversion, abuse and unintended exposure to Bup. RBP-6000 is currently being assessed for efficacy and safety in a phase 3 clinical trial.